Poster A1

Poster Session A: Prostate Cancer, February 17, 2022, 11:30–13:00; 17:45–18:45
Rapid Abstract Session: Prostate Cancer, February 17, 2022, 16:45–17:45

Presenting author: Alicia K Morgans
Email: aliciak_morgans@dfci.harvard.edu

Identifying patient profiles and mapping the patient journey across 3 countries in a large-scale, fully digital survey of patients with prostate cancer

Alicia K Morgans,[1] Reiner Lehmann,[2] Axel Heidenreich,[3] Stephen Allen,[4] Ernst-Günther Carl,[5] Howard Wolinsky,[6] Andreas Poschenrieder,[7] Osvaldo Mirante,[7] Joe M O'Sullivan[8]

[1]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [2]DontBePatient Intelligence GmbH, Munich, Germany; [3]Department of Urology, University of Cologne, Cologne, Germany; [4]Tackle Prostate Cancer, London, UK; [5]Europa Uomo, Antwerp, Belgium; [6]AnCan/UsToo Virtual Support Group for Active Surveillance, Flossmor, IL, USA; [7]Advanced Accelerator Applications, a Novartis company, Geneva, Switzerland; [8]Queen's University Belfast, Belfast, Northern Ireland

Scan to obtain:
- **Poster**
- **Slides**

https://bit.ly/AliciaK1
Copies of this poster and oral presentation slides obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- To our knowledge, this is the largest digital survey to date conducted in patients with prostate cancer, providing insights into areas of potential improvement from distinct patient populations by disease state and country.
- Preliminary data from this patient-reported data set suggest that rates of prostate cancer detection by screening may be lower in the UK than the US and Germany.
- Different treatment patterns between countries may reflect differences in healthcare systems and the professional teams involved.
- Active surveillance rates were relatively lower in the US and Germany compared with the UK, where this therapeutic strategy is more common.
- In patients with non-metastatic disease, those who completed the survey in the US and Germany more commonly reported prostatectomy as the local treatment used, whereas radiotherapy was more commonly reported by patients completing the survey in the UK.
- Further analyses will include the impact of differences in QoL outcomes, patient journey, trust in healthcare professionals, access to information, and involvement with patient advocacy groups.

Funded by Advanced Accelerator Applications, a Novartis company.

Poster presented at the American Society of Clinical Oncology Genitourinary Cancers Symposium, held on February 17–19, 2022.

INTRODUCTION

- Prostate cancer is the second most frequently occurring cancer in men worldwide.[1]
- Mortality has decreased in Western countries since the mid-1990s because of advances in therapy and diagnosis,[2,3] which means that patients with prostate cancer are living longer.
- Patients' experiences in terms of the care they receive and their quality of life (QoL) are important for decision-making, as well as long-term satisfaction.[4–6]
- We conducted the largest multinational digital survey to date, in terms of both scope and number of patients with prostate cancer.
 - Previous large-scale surveys assessing the prostate cancer patients' journey were largely limited to one country and/or did not utilize digital technology.[7–9]

Objective

- We designed and distributed a digital survey online, to capture the patient journey across the prostate cancer disease continuum (including both non-metastatic and metastatic prostate cancer), from the time of diagnosis.
- We sought to map patients' experiences, expectations, and attitudes, and identify challenges and unmet needs in diagnosis, therapy patterns, care teams, QoL, patient organizations, and resources.

METHODS

- This novel digital survey was designed by DontBePatient Intelligence, in collaboration with patient organizations and medical experts from the US, UK and Germany.
- The survey comprised an average of 83 questions. Patients had the option of completing the survey at one sitting or pausing and continuing later.
 - The total number of questions was based on the number of therapies received, with each therapy linked to specific, detailed follow-up questions.
 - Participants who received chemotherapy were prompted to complete additional questions related to the number of agents included in the regimen, as well as number of cycles received.
 - Participants who confirmed that they experienced pain were prompted to complete the Brief Pain Inventory (current version, revision 07/01/05).[10]
- Patients/carers were recruited through social media advertising (Facebook advertising, Google search engine marketing, and network banners) and patient organizations (link sharing on their website) to avoid institutional and organizational bias.
- The study opened for recruitment on February 9, 2021; the recruitment goal was reached in 60 days, on April 10, 2021, at which point the study was closed.
- Data analysis was performed using predominantly descriptive methods and inductive statistics (Pearson's Chi-squared test) where applicable.

RESULTS

Demographics

- The campaigns generated 610,378 landing page visits and 33,882 survey respondents, of which 15,824 completed the survey.
- Participants were equally distributed between the three participating countries, with similar contribution from rural and urban areas (**Table 1**).
 - The baseline characteristics were well balanced between patients with non-metastatic (M–) and metastatic (M+) disease.
 - No between-country differences in median age (70.0 ± 7.2 years) and median Gleason score (7.0 ± 1.5) were observed.
 - ~80% of completers had M– disease at the time of survey completion.
 - The mean number of therapies was greater for patients with M+ disease (2.1) than those with M– disease (1.2).
 - The proportion of patients on active surveillance was highest in the UK (11.5%, vs 7.4% and 5.7% in the US and Germany, respectively).

Table 1. Patient demographics by country

	US	UK	Germany
Completers, n (%)	5,548 (35.1%)	5,397 (34.1%)	4,879 (30.8%)
M–	4,594 (29.0%)	4,260 (26.9%)	3,950 (25.0%)
M+	954 (6.0%)	1,137 (7.2%)	929 (5.9%)
Median age, years	69.0	70.0	70.0
M–	69.0	70.0	70.0
M+	69.0	70.0	70.0
Median Gleason score	7.0	7.0	7.0
M–	7.0	7.0	7.0
M+	8.0	8.0	8.0
Mean number of therapies (M–), n	1.4	1.1	1.3
Mean number of therapies (M+), n	2.2	2.0	2.0
Proportion of patients on active surveillance, %	7.4%	11.5%	5.7%
Type of residence	Rural: 50.1% / Urban: 49.9%		

M–, non-metastatic disease; M+, metastatic disease

Initial mode of diagnosis

- The most common initial mode of diagnosis (active screening vs incidental vs symptomatic) differed between countries (**Figure 1**).

Figure 1. Initial mode of diagnosis by country and disease stage

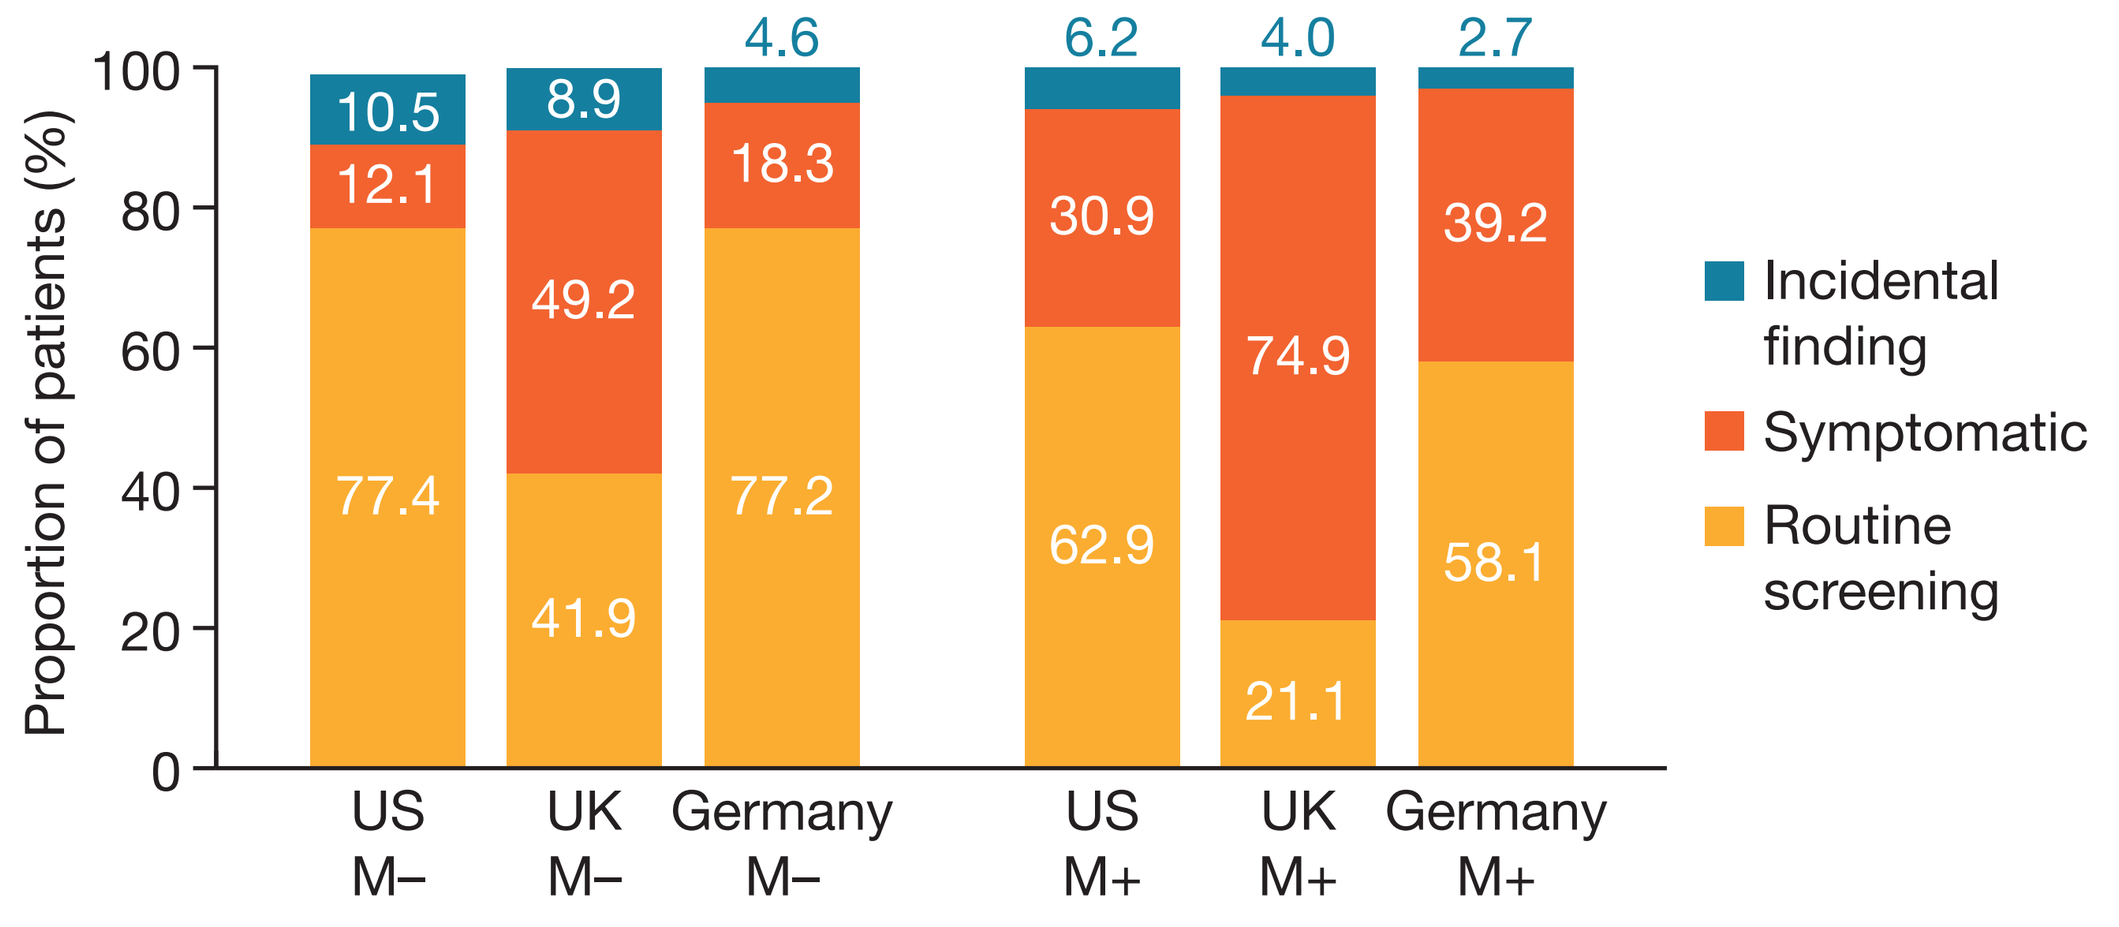
Proportion of patients (%)
0
20
40
60
80
100
10.5
12.1
77.4
8.9
49.2
41.9
4.6
18.3
77.2
6.2
30.9
62.9
4.0
74.9
21.1
2.7
39.2
58.1
US M–
UK M–
Germany M–
US M+
UK M+
Germany M+
Incidental finding
Symptomatic
Routine screening

M–, non-metastatic disease; M+, metastatic disease

- In the US and Germany, diagnosis through healthcare screening was more frequent than in the UK (M–/M+: US 77.4%/62.9%; UK 41.9%/21.1%; Germany 77.2%/58.1%), where 74.9% of patients with M+ disease had a symptomatic diagnosis.
- In all three countries, patients with M+ disease had a symptomatic diagnosis more frequently than those with M– disease (M–/M+: US 12.1%/30.9%; UK 49.2%/74.9%; Germany 18.3%/39.2%).

Treatment patterns

- Statistically different treatment patterns were observed in the three participating countries ($p < 0.001$), although the effect size is relatively small (phi = 0.221).
- In the overall population, approximately half of the patients (US 53.1%; UK 48.2%; Germany 60.3%) reported receiving only one prior treatment at the time of survey completion.
- Prostatectomy was the most commonly used local therapy for M– disease in the US (57.4%) and Germany (71.3%), followed by radiotherapy and hormone therapy; in the UK, radiotherapy (47.6%) was slightly more frequently used than prostatectomy and hormone therapy (**Figure 2**).
 - Across the three participating countries, fewer than 1.5% of patients received chemotherapy for M– disease.

Figure 2. Types of therapies received by country and disease stage

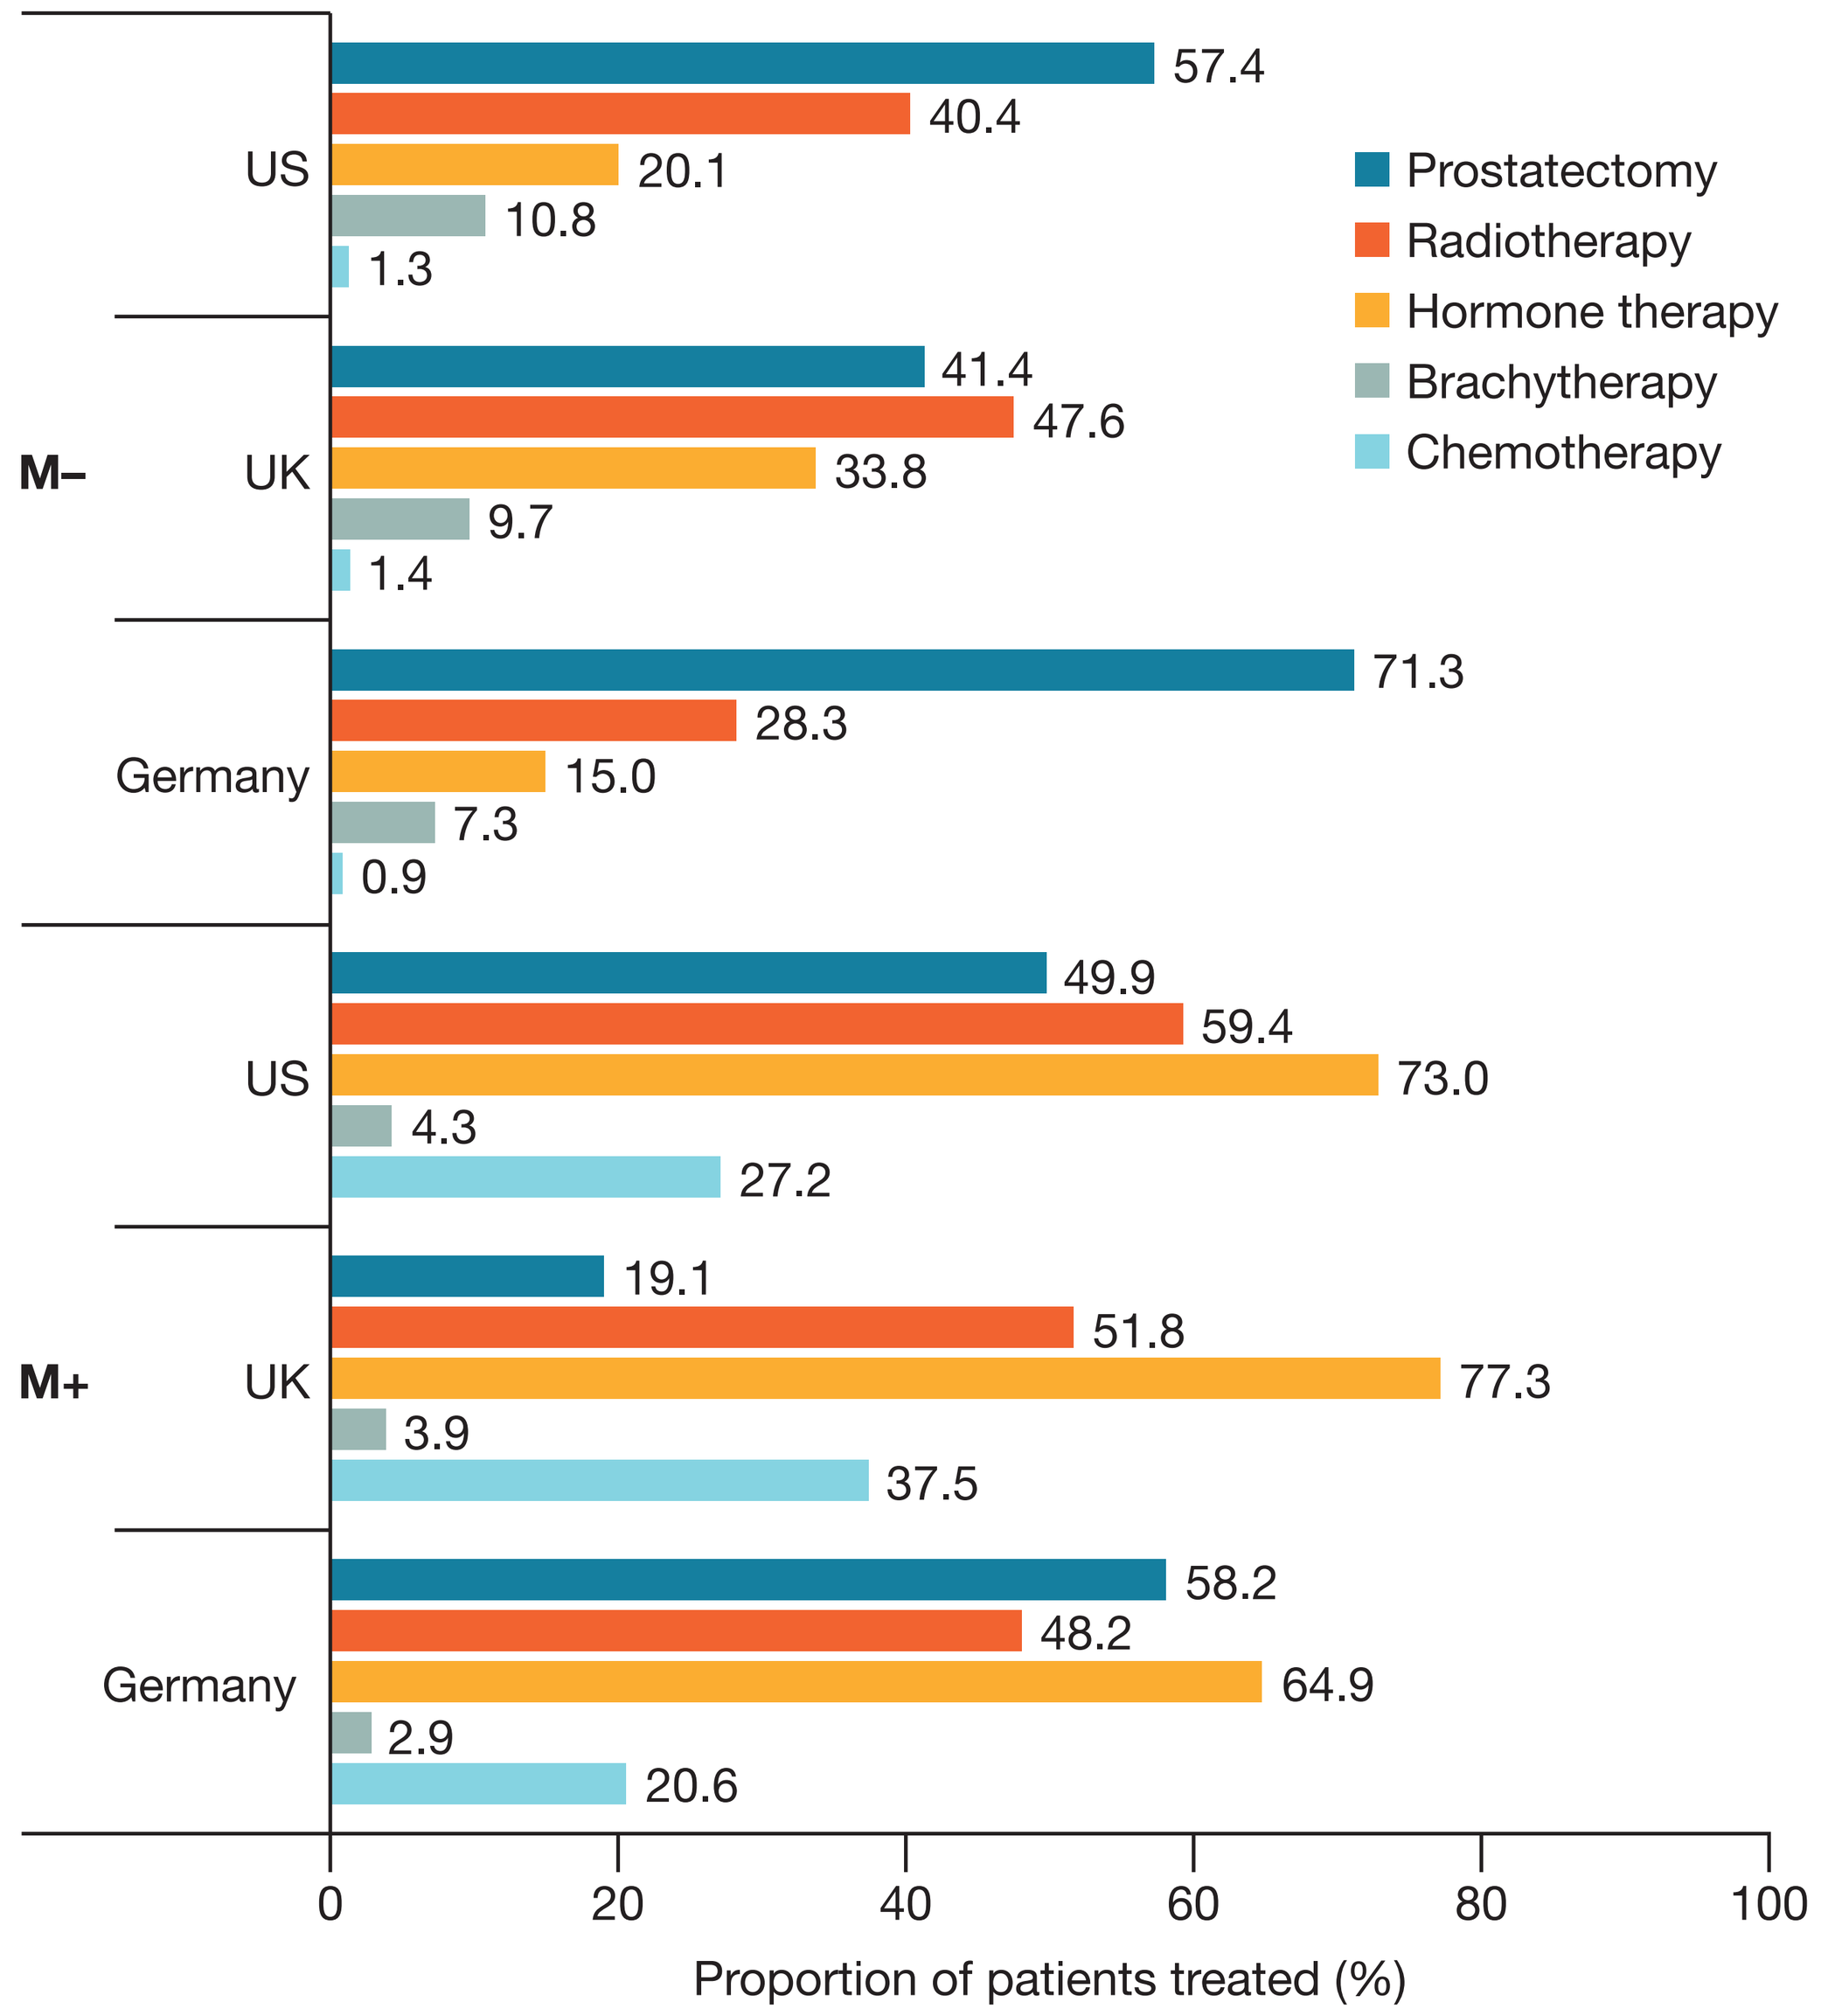
M–
US
57.4
40.4
20.1
10.8
1.3
UK
41.4
47.6
33.8
9.7
1.4
Germany
71.3
28.3
15.0
7.3
0.9
M+
US
49.9
59.4
73.0
4.3
27.2
UK
19.1
51.8
77.3
3.9
37.5
Germany
58.2
48.2
64.9
2.9
20.6
0
20
40
60
80
100
Proportion of patients treated (%)
Prostatectomy
Radiotherapy
Hormone therapy
Brachytherapy
Chemotherapy

M–, non-metastatic disease; M+, metastatic disease
Hormone therapy included androgen receptor pathway inhibitors and androgen deprivation therapy
This graphic does not reflect the sequence of treatment received

- For M+ disease, hormone therapy was the most common therapy (US 73.0%; UK 77.3%; Germany 64.9%) in all countries (**Figure 2**).
 - Chemotherapy was more frequently used in the UK (37.5%) than in the US (27.2%) and Germany (20.6%).
- Overall, of the M– and M+ patients who received chemotherapy, the majority (≥ 65.5%) received one type of chemotherapy in all countries (**Figure 3**), and most patients did not remember the name(s) of the chemotherapies received.
 - Of the patients who remembered the name of the chemotherapy agents received, ~65% received taxane-based chemotherapy.
- A higher percentage of patients with M– disease were on active surveillance in the UK (14.2%) compared with the US (8.7%) and Germany (6.4%) (**Figure 4**).

Figure 3. Number of chemotherapy regimens received by patients

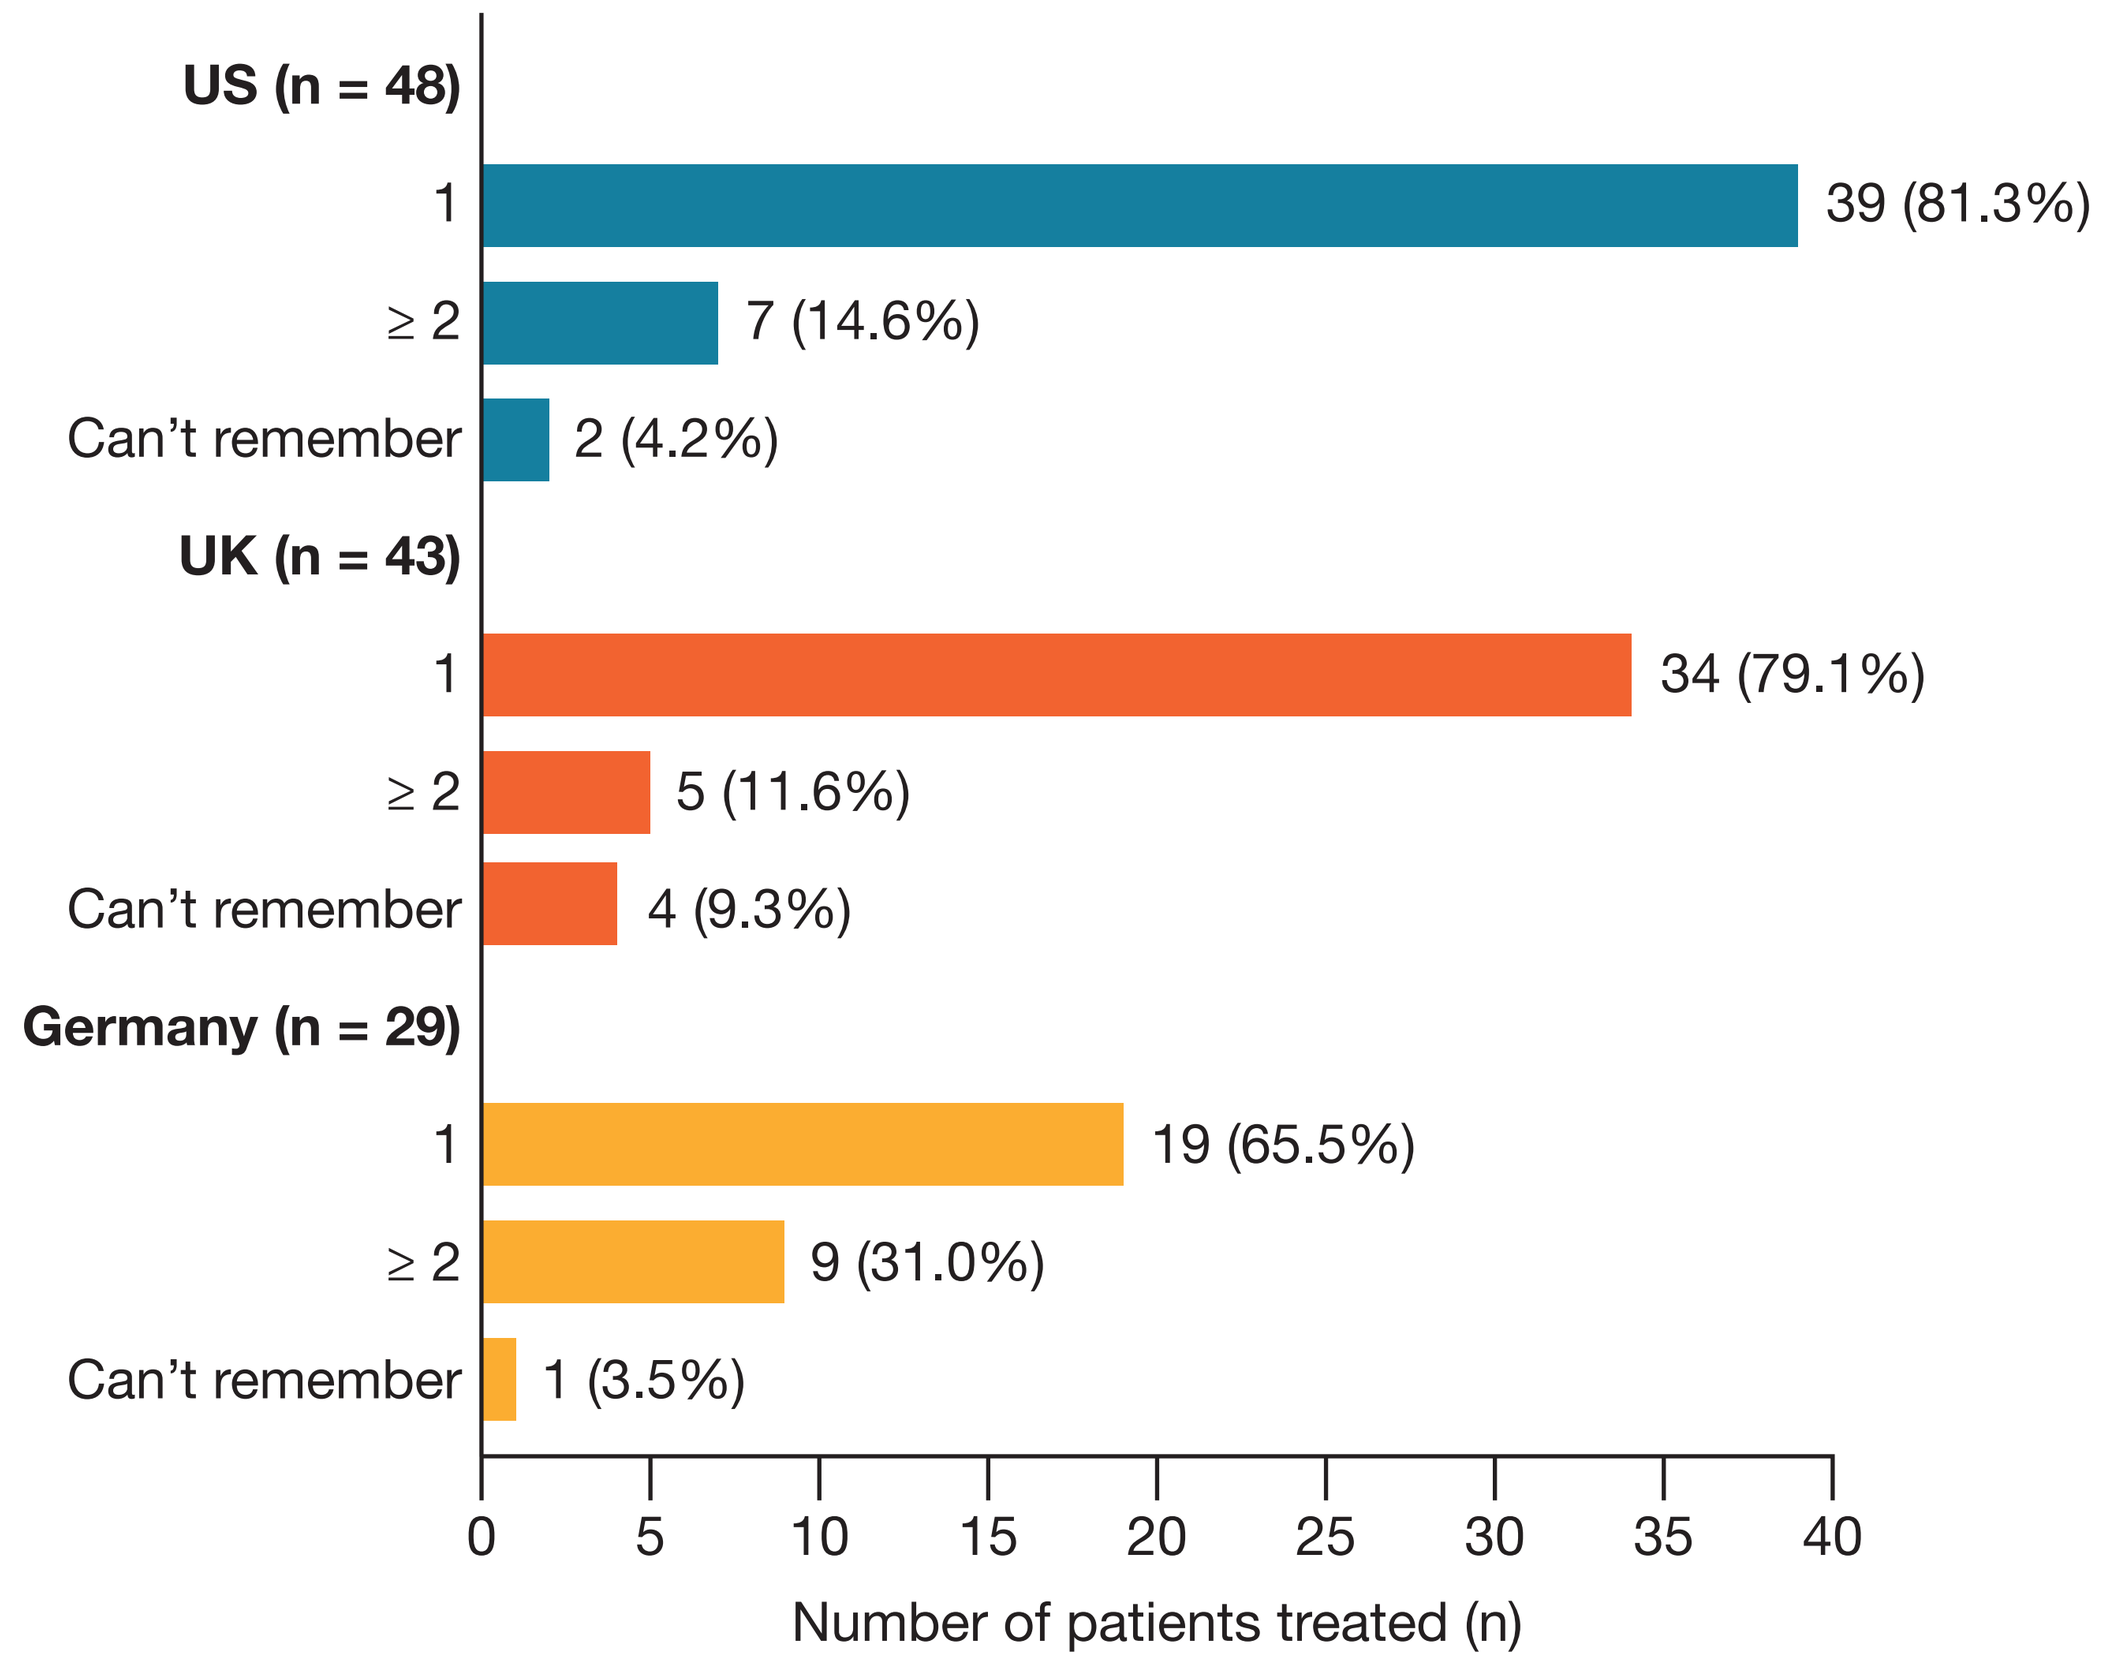
US (n = 48)
1
39 (81.3%)
≥ 2
7 (14.6%)
Can't remember
2 (4.2%)
UK (n = 43)
1
34 (79.1%)
≥ 2
5 (11.6%)
Can't remember
4 (9.3%)
Germany (n = 29)
1
19 (65.5%)
≥ 2
9 (31.0%)
Can't remember
1 (3.5%)
0
5
10
15
20
25
30
35
40
Number of patients treated (n)

1 = one type of chemotherapy; ≥ 2 = two or more types of chemotherapy

Figure 4. Proportion of patients on active treatment vs active surveillance in patients with M– disease by country

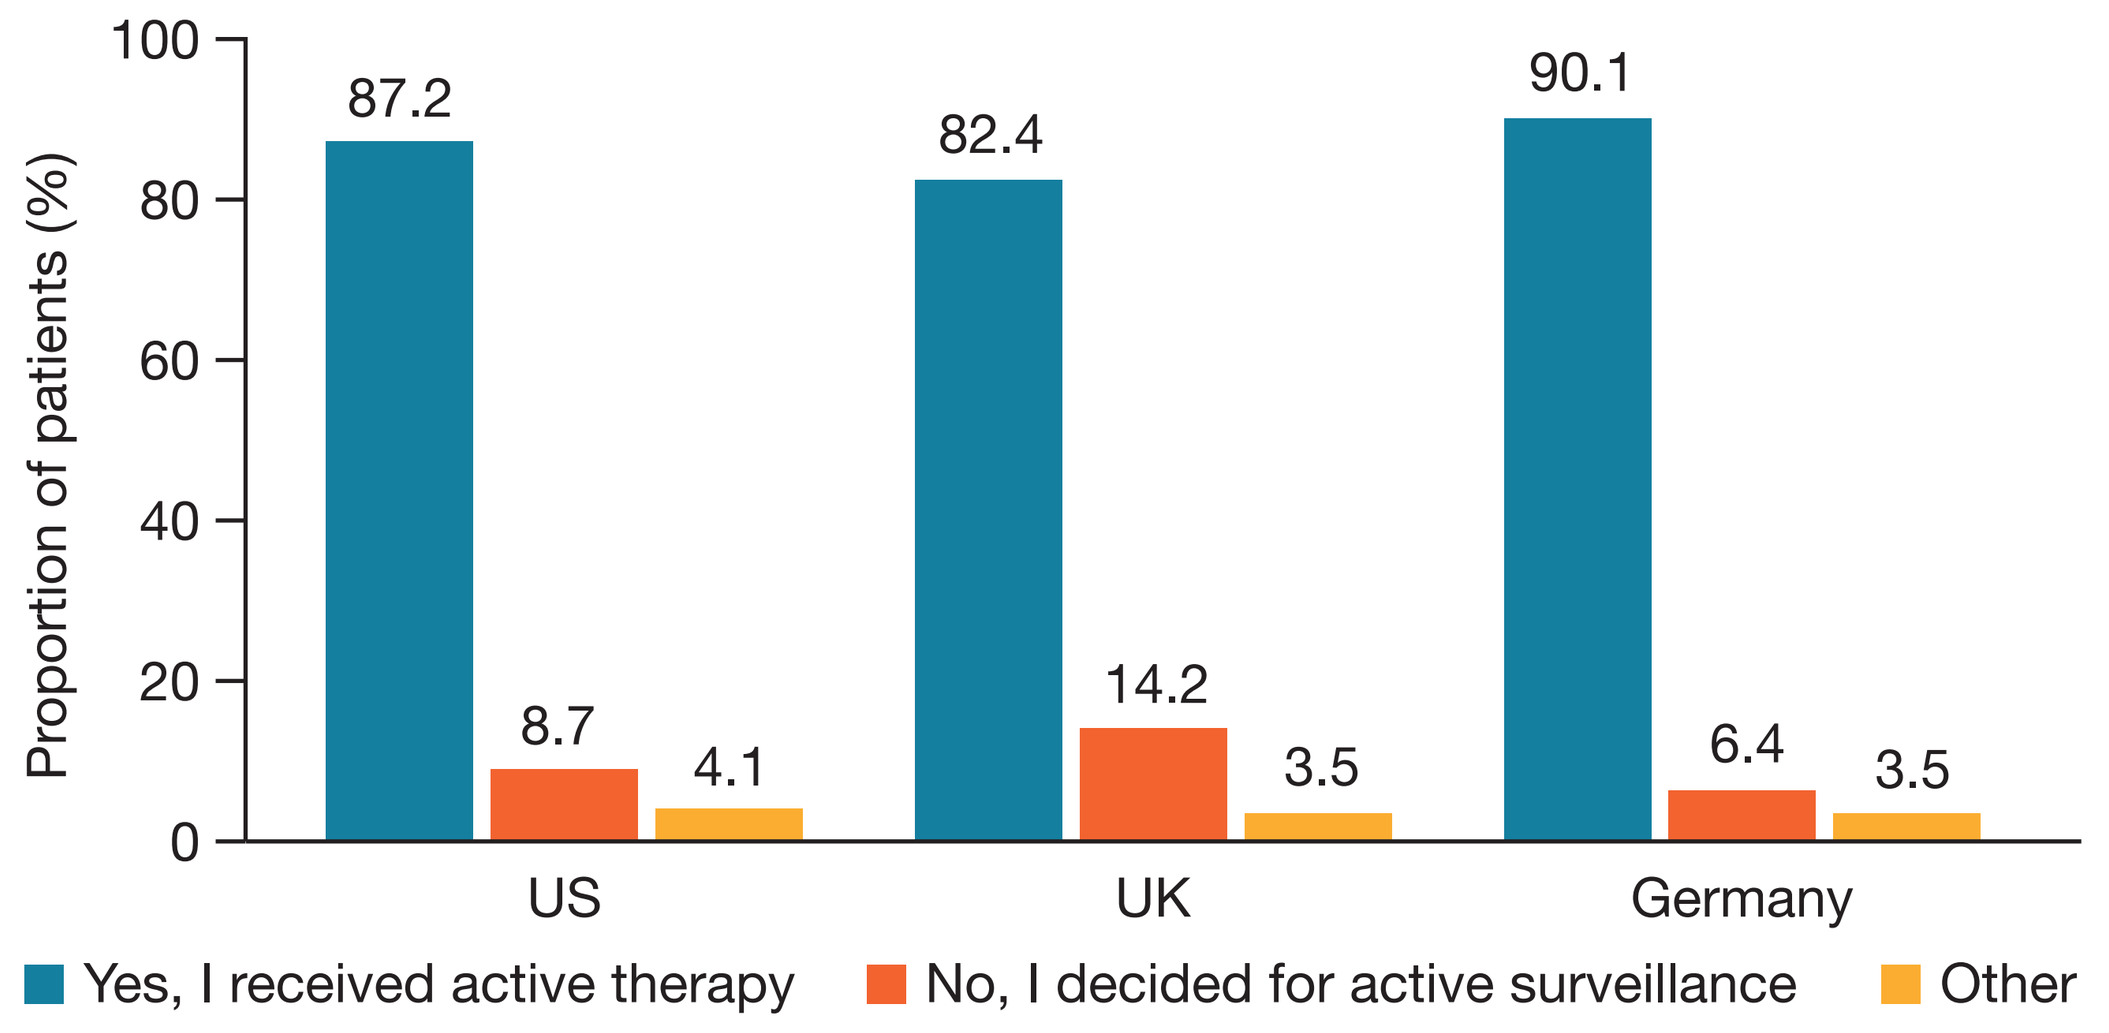
Proportion of patients (%)
0
20
40
60
80
100
87.2
8.7
4.1
82.4
14.2
3.5
90.1
6.4
3.5
US
UK
Germany
Yes, I received active therapy
No, I decided for active surveillance
Other

Patient expectation of therapy received

- In patients who were not receiving active therapy at the time of survey, all treatments were viewed relatively negatively, including hormonal therapy (data not shown).
 - Chemotherapy was perceived negatively in Germany and the US, and more positively in the UK.
- In total, 85.3% of patients in the US perceived prostatectomy to be an effective type of therapy.
 - More than 85.5% of patients in the UK perceived prostatectomy and radiotherapy as effective, and 83.6% and 82.6% of German patients perceived radiotherapy and active surveillance, respectively, as an effective type of therapy (**Figure 5**).

Figure 5. Patient ratings of expected efficacy of treatment

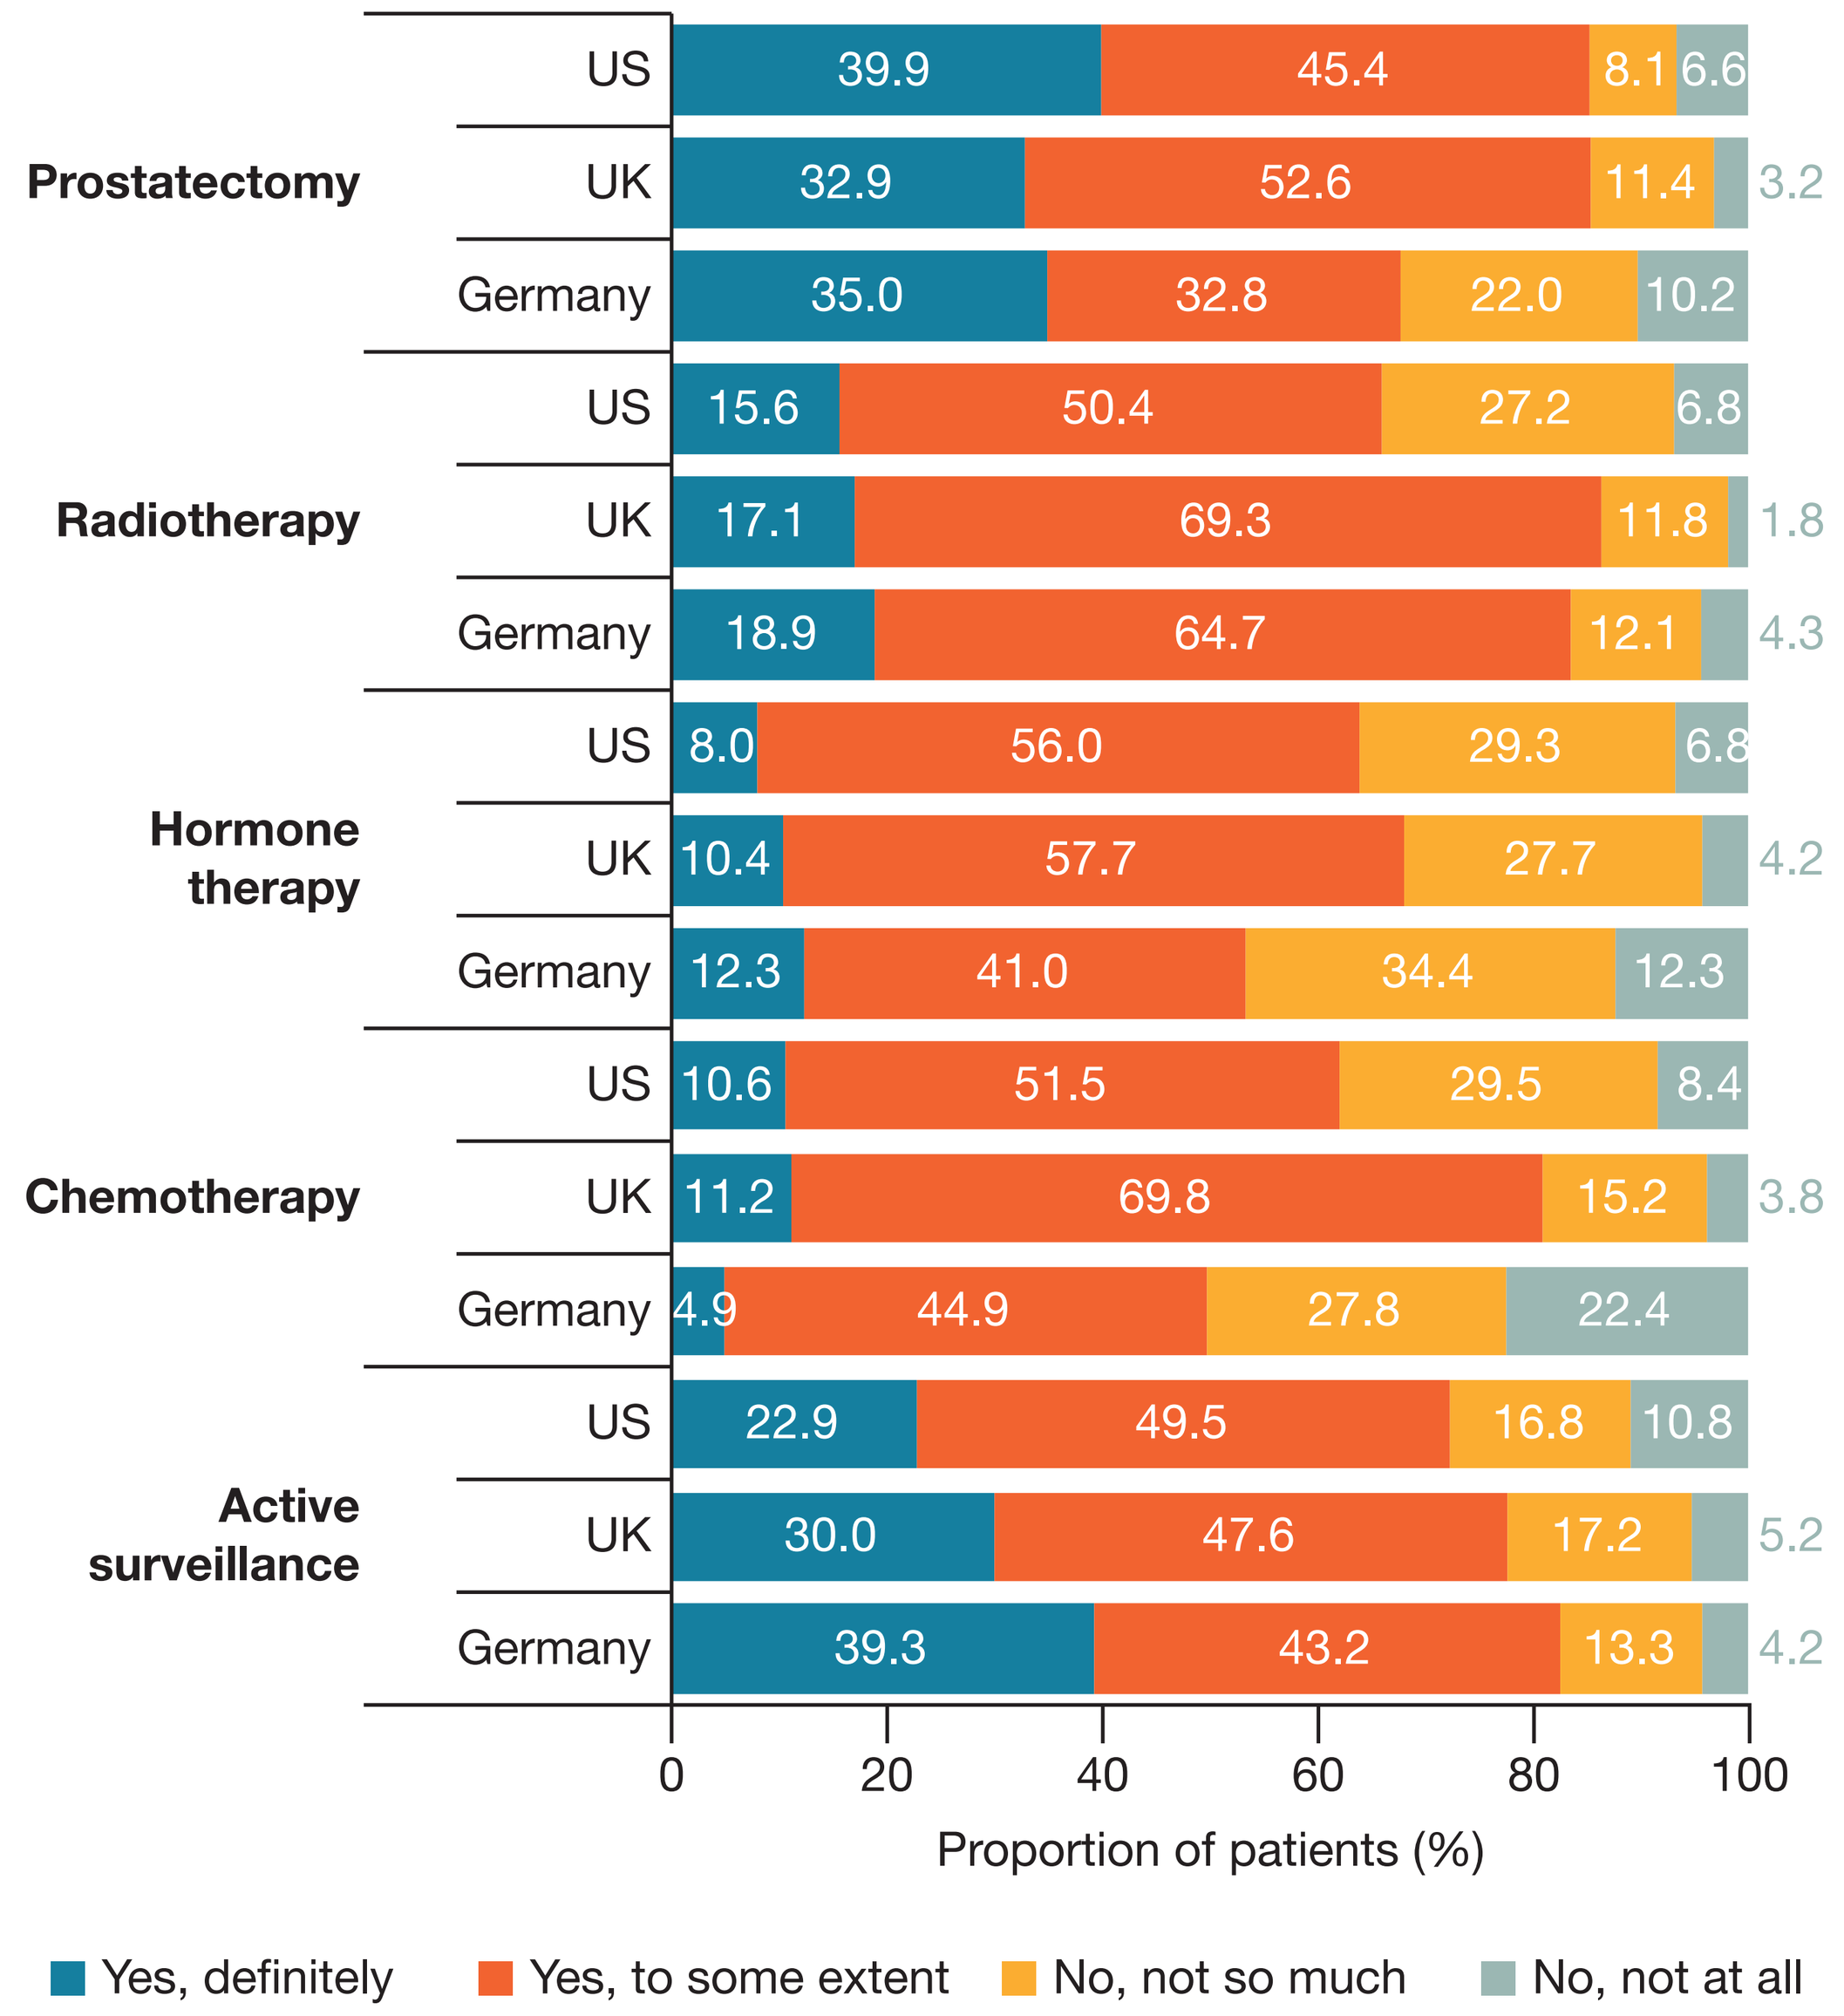
Prostatectomy
US 39.9 45.4 8.1 6.6
UK 32.9 52.6 11.4 3.2
Germany 35.0 32.8 22.0 10.2
Radiotherapy
US 15.6 50.4 27.2 6.8
UK 17.1 69.3 11.8 1.8
Germany 18.9 64.7 12.1 4.3
Hormone therapy
US 8.0 56.0 29.3 6.8
UK 10.4 57.7 27.7 4.2
Germany 12.3 41.0 34.4 12.3
Chemotherapy
US 10.6 51.5 29.5 8.4
UK 11.2 69.8 15.2 3.8
Germany 4.9 44.9 27.8 22.4
Active surveillance
US 22.9 49.5 16.8 10.8
UK 30.0 47.6 17.2 5.2
Germany 39.3 43.2 13.3 4.2
0
20
40
60
80
100
Proportion of patients (%)
Yes, definitely
Yes, to some extent
No, not so much
No, not at all

Hormone therapy included androgen receptor pathway inhibitors and androgen deprivation therapy

Patient satisfaction with care teams involved in treatment management

- Satisfaction levels were generally high in all countries, at more than 80% for the care teams involved in the application of all therapies.
- Differences in satisfaction levels with care teams between therapies were low, with no observed link to treatment outcome.
- There was a trend for decrease in patient satisfaction over time, from time of diagnosis, independent of therapy type (data not shown).

Acknowledgments

The authors thank the patients who participated in this study. Under direction of the authors, Shufei Song, PhD, from Oxford PharmaGenesis, Oxford, UK, provided medical writing support, which was funded by Advanced Accelerator Applications, a Novartis company, in accordance with Good Publication Practice (GPP3) guidelines (https://www.ismpp.org/gpp3).

Disclosures

Alicia K Morgans reports consultancy fees from Astellas, AstraZeneca, AAA, Bayer, Clovis, Dendreon, Exelixis, Janssen, Lantheus, Merck, Myovant, Myriad, Novartis, Pfizer, Sanofi, Seattle Genetics, contracted research from Astellas, Bayer, Myovant, Seattle Genetics, and honoraria from Astellas, AstraZeneca, Janssen, Myovant, Pfizer, Sanofi

References

1. Sung H *et al. CA Cancer J Clin* 2021;71:209–49. **2.** Etzioni R *et al. Cancer Causes Control* 2008;19:175–81. **3.** Tsodikov A *et al. Ann Intern Med* 2017;167:449–55. **4.** Resnick MJ *et al. N Engl J Med* 2013;368:436–45. **5.** Sanda MG *et al. N Engl J Med* 2008;358:1250–61. **6.** Shevach J *et al. Curr Urol Rep* 2019;20:57. **7.** The Royal College of Surgeons of England. NPCA Annual Report 2018. Available from: https://www.npca.org.uk/content/uploads/2019/02/NPCA-Annual-Report-2018.pdf. (Accessed January 2022). **8.** Nguyen-Nielsen M *et al. Cancer Epidemiol* 2020;64:101623. **9.** Lehto US *et al. Eur J Oncol Nurs* 2015;19:220–9. **10.** Cleeland CS. Brief Pain Inventory (Short Form). 07/01/05 Current version. Available from: http://www.npcrc.org/files/news/briefpain_short.pdf. Accessed January 2022.